Speech phenotypes in cognitively healthy participants at risk of developing Alzheimer's disease

Jennifer Sorinas[1], Jessica Robin[2], William Simpson[2,3], Jelena Curcic[1], Kristin Hannesdottir[4]
[1]Novartis Institutes for Biomedical Research, Basel, Switzerland
[2]Winterlight Labs, Inc. Toronto, Canada
[3]McMaster University, Hamilton, Canada
[4]Novartis Institutes for Biomedical Research, Cambridge, MA, United States

Background

- Changes to speech patterns have been identified as early signs of Alzheimer's Disease (AD) and have been shown to progress with disease.[1,2,3,4]
- It is not known if changes to speech that occur prior to any detectable cognitive decline can be used to identify risk for AD.
- The objective of this project was to use data-driven approaches to identify speech phenotypes in a sample of cognitively healthy participants at risk of developing AD.

Methods

- We analyzed baseline Clinical Dementia Rating (CDR) interview recordings from 114 participants (66% women; age range = 59-76, Table 1).
- Participants were cognitively healthy but had risk factors for developing AD (APOE4+ and Aβ+).
- CDR recordings were segmented, diarized and transcribed and 8 categories of speech features were extracted using the Winterlight speech analysis platform (Figure 1).
- After data preprocessing, cleaning and standardization, we performed a dimensionality reduction analysis (PCA) within each feature category and extracted the first two components.
- Blinded k-means cluster analysis was performed (n clusters = 2) to determine if participants clustered into subgroups based on speech features.

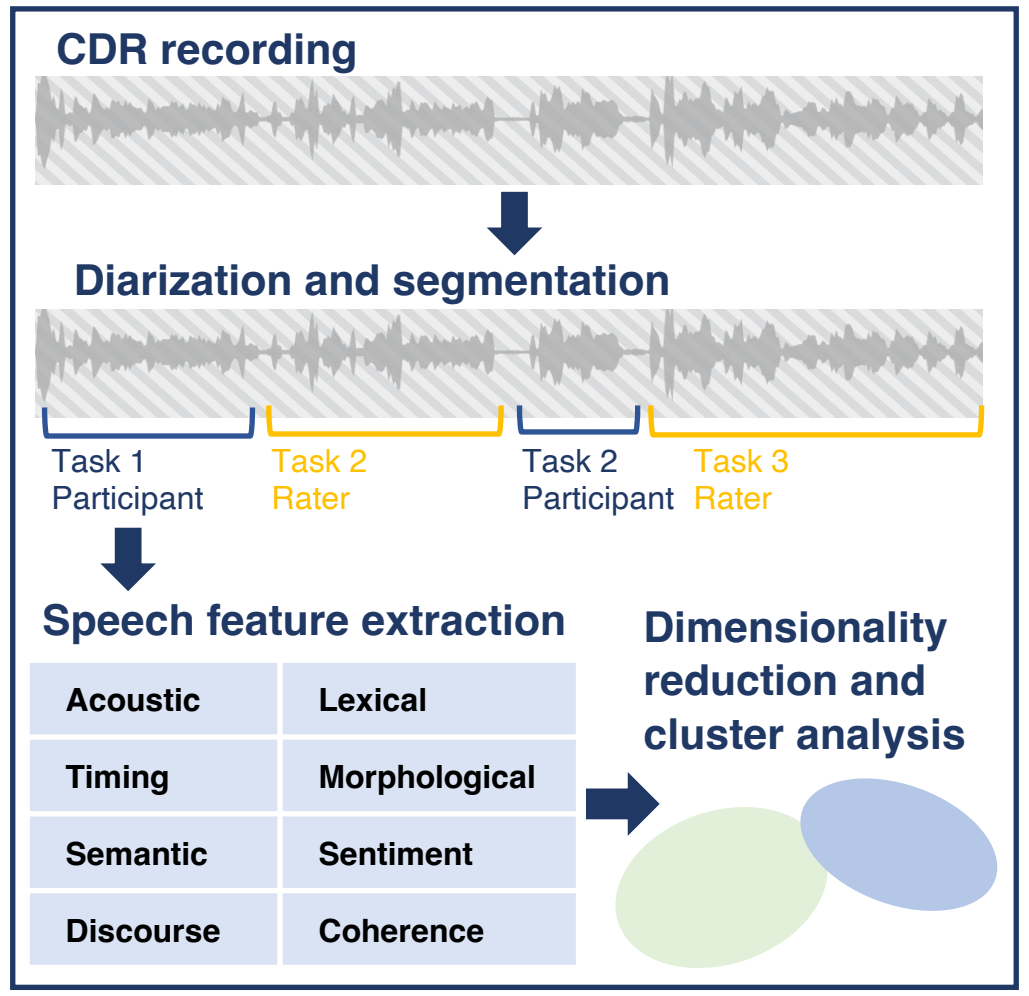
CDR recording
Diarization and segmentation
Task 1
Participant
Task 2
Rater
Task 2
Participant
Task 3
Rater
Speech feature extraction
Acoustic
Lexical
Timing
Morphological
Semantic
Sentiment
Discourse
Coherence
Dimensionality reduction and cluster analysis

Figure 1. Schematic of study methods

N	Age, mean (SD)	Sex, n male (%)
114	68.6 (4.4)	39 (34%)

Table 1. Baseline sample demographics.

Results

- Silhouette analysis yielded two clusters of participants who differed on the timing and acoustic feature categories, in the "address repeat" and "recent experience" sections of the CDR interview.
- One cluster (blue; Figure 2) showed a significant increase in average word duration, higher hesitations, more filled pauses, and longer audio duration in the "address repeat" section.

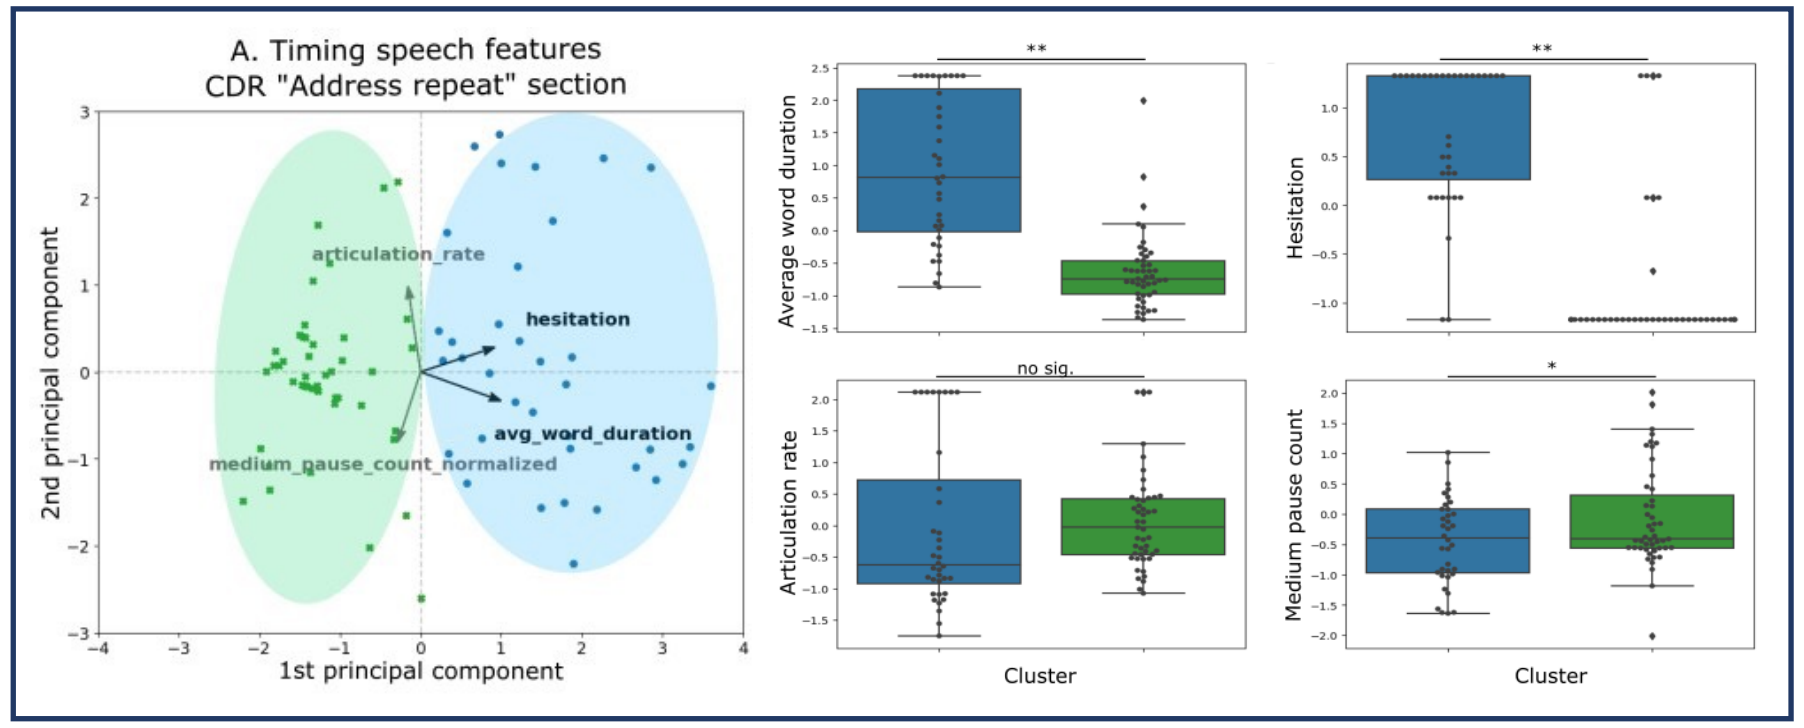
A. Timing speech features
CDR "Address repeat" section
articulation_rate
hesitation
avg_word_duration
medium_pause_count_normalized
2nd principal component
1st principal component
Average word duration
Hesitation
Articulation rate
Medium pause count
**
**
no sig.
*
Cluster
Cluster

Figure 2. Cluster analysis and selected speech features from the "address repeat" section of the CDR.

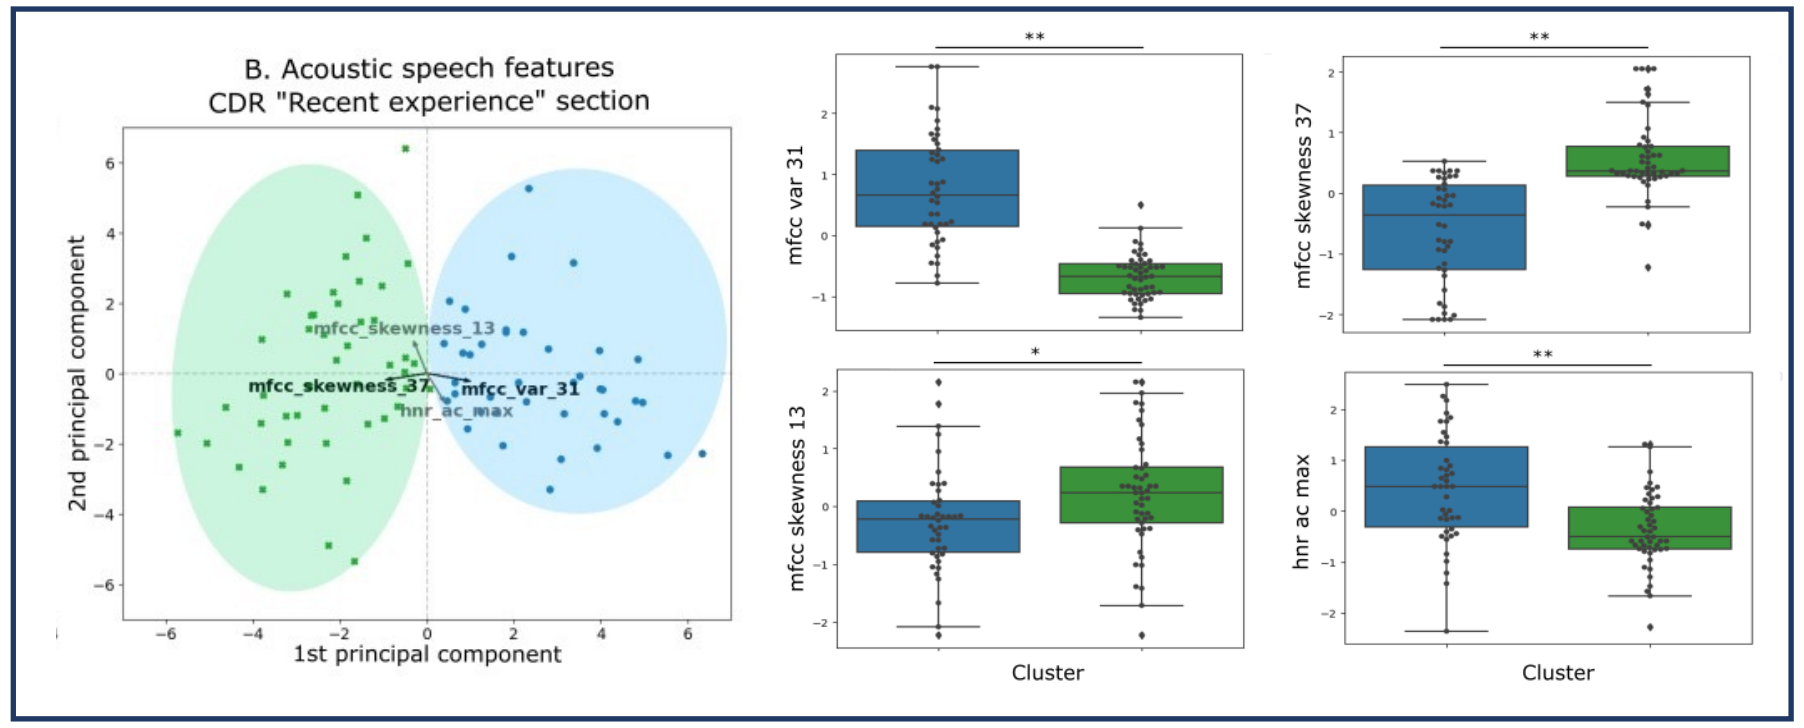
B. Acoustic speech features
CDR "Recent experience" section
mfcc_skewness_13
mfcc_skewness_37
mfcc_var_31
hnr_ac_max
2nd principal component
1st principal component
mfcc var 31
mfcc skewness 37
mfcc skewness 13
hnr ac max
**
**
*
**
Cluster
Cluster

Figure 3. Cluster analysis and selected speech features from the "recent experience" section of the CDR.

- The groups also differed on acoustic features in the "recent experience" section (Figure 3). These results suggest that the group of participants who struggled more with the "address repeat" item also present a different speech acoustic phenotype.
- These clusters did not differ on conventional clinical endpoint scores, such as RBANS and MMSE, indicating that speech measures may detect subgroups at preclinical stages of AD that do not differ on cognitive assessments.

Results (continued)

- The speech-based clusters did not differ on additional variables, including age, sex, BMI, APOE4 status, and hippocampal volume.

Conclusions

- This project demonstrates how data-driven methods can identify speech phenotypes from naturalistic, passively collected, speech recordings.
- Cluster analysis indicated that at baseline, before any sign of cognitive decline, participants could be distinguished into two groups based on timing and acoustic parameters of speech.
- These different patterns of speech did not correspond to scores on conventional clinical endpoints, demographic variables or AD risk factors.
- More research is needed to understand how speech phenotypes relate to disease progression and correlate with other clinical measures and AD biomarkers.

References

1. Robin, J., Xu, M., Kaufman, L. D. & Simpson, W. Using Digital Speech Assessments to Detect Early Signs of Cognitive Impairment. Front. Digit. Health 3, 749758 (2021).
2. Eyigoz, E., Mathur, S., Santamaria, M., Cecchi, G. & Naylor, M. Linguistic markers predict onset of Alzheimer's disease. EClinicalMedicine 28, 100583 (2020).
3. Amini, S. et al. Automated detection of mild cognitive impairment and dementia from voice recordings: A natural language processing approach. Alzheimers Dement. alz.12721 (2022) doi:10.1002/alz.12721.
4. König, A. et al. Automatic speech analysis for the assessment of patients with predementia and Alzheimer's disease. Alzheimers Dement. Diagn. Assess. Dis. Monit. 1, 112–124 (2015).

Copyright © 2023 - Contact: jennifer.sorinas_nerin@novartis.com